3583

Dose escalation and pharmacodynamic study of IV REOLYSIN®, a proprietary formulation of a live replication competent RNA virus in patients with advanced solid tumors

M. H. Ghalib[1], K.K.Desai[1], R. Gollamudi[1], I. Chaudhary[1], M. Einstein[1], M. Coffey[2], K. Mettinger[2], S. Mani[1], S. Goel[1];
[1]Montefiore Medical center, Bronx, NY; [2]Oncolytics Biotech Inc., Calgary, AB, Canada

ABSTRACT

Background: Reolysin® is a proprietary formulation of reovirus serotype 3 - Dearing strain, a double-stranded replication-competent RNA non-enveloped icosahedral virus. It induces cytopathic/anti-cancer effects in cells with an active Ras pathway due to ras mutation or upstream activation (e.g. EGF expression/mutation).

Methods: This was a single center dose escalation trial of Reolysin administered IV in doses ranging from single doses of 1X10^8 to multiple doses every 4 weeks of 3X10^{10} tissue culture infective dose (TCID)$_{50}$. Serum for neutralizing antibody, and serum, stool, saliva, and urine for viral shedding were collected. Tumor samples were analyzed for activating mutations in the ras and b-raf oncogenes.

Results: Eighteen patients received 27 doses of Reolysin in 6 dose cohorts accomplishing a 300 fold dose escalation without a protocol-defined dose limiting toxicity. Drug related grade 2 toxicities included fatigue and fever (1 patient each). All patients developed neutralizing antibodies during the course of the study. Viral shedding was observed in 6 patients [urine (5), serum (4), saliva (3), and stool (2)]. One patient with anthracycline and taxane refractory breast cancer experienced a partial response (PR) and her tumor had a mutation in the ras oncogene. Biopsy from a chest wall mass showed evidence of necrosis and viral replication by electron microscopy. The overall clinical benefit rate was 45% and appeared higher in patients with viral shedding (67%) than those without (33%).

Conclusion: Reolysin administered as a one hour infusion on a monthly schedule is safe and well-tolerated even in multiple doses. Reolysin has anti-tumor activity as a single agent warranting further evaluation, including in combination with chemotherapy. Viral shedding may suggest intrapatient replication yielding a benefit and should be studied carefully in future studies.

BACK GROUND & MECHANISM OF ACTION

◆REOLYSIN – a proprietary formulation of reovirus, a naturally occurring, replication competent virus
◆Typically asymptomatic in humans (does not cause disease)
◆Replicates in Ras-activated cancer cells resulting in cell death
◆ Fully replication-competent
◆Mamalian permissive therefore effective modeling in murine and non-human primates
◆Replication is exclusively cyctoplasmic
◆Proof of viral replication in tumors following systemic delivery

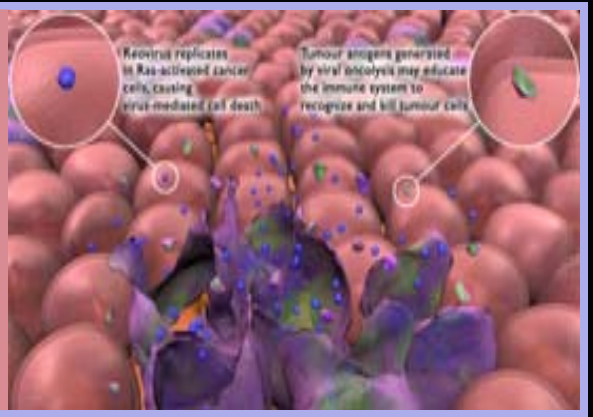
Reovirus replicates in Ras-activated cancer cells, causing virus-mediated cell death
Tumour antigens generated by viral oncolysis may educate the immune system to recognize and kill tumour cells

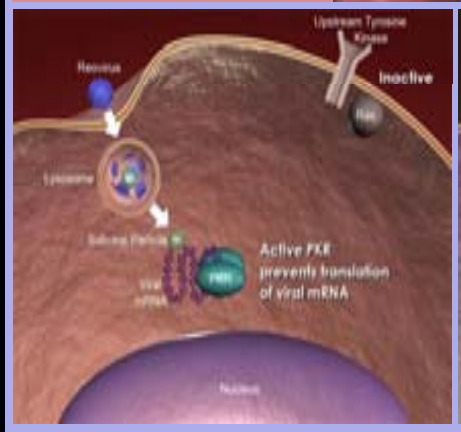
Upstream Tyrosine Kinase
Inactive
Reovirus
Lysosome
Active PKR prevents translation of viral mRNA

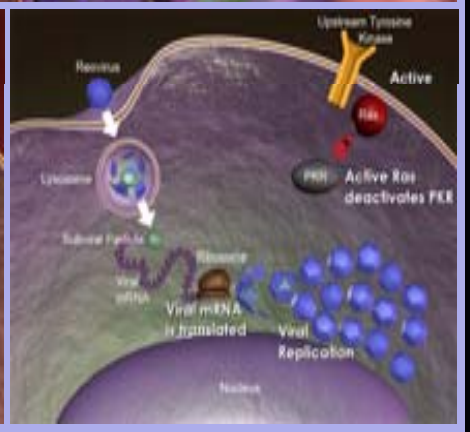
Upstream Tyrosine Kinase
Active
Reovirus
Lysosome
Active Ras deactivates PKR
Viral mRNA is translated
Viral Replication

ANALYSIS

BASELINE PATIENT CHARACTERISTICS

Sex	*Female*	14 (78%)
	Male	4 (22%)
	Total	18
Age	*Range*	40-72
Tumor types	*Ovary*	6
	Colon	4
	Breast	2
	Others	6
Prior chemotherapy regimens	*1-3*	6
	4-8	8
	> 9	4
Total doses	*Total*	27
	Median	1(1-7)
	3 pts multiple cycles	2,3,7 doses
Performance status	*1*	17
	2	1

METHODODOLOGY

Design	Standard phase I dose escalation
Dose(TCID$_{50}$)	1X10^8 - 3X10^{10} TCID$_{50}$ (300 X)
Administration	Initially single dose q 4 wks, then multiple doses
Infusion	1 hr IV infusion in 250 cc
Safety	Precautions for patient and family
CT/MUGA	CT/MUGA at 0,1,3,5,7 months
HbsAg/HIV	Negative

SINGLE PATIENT PROFILE

The post treatment biopsy revealed extensive necrosis in the dermis, well demonstrated on the standard hematoxylin and eosin staining .

Age/Sex/Race	60/F/White
Primary	Breast Ca. refractory to taxanes & anthracyclines
Response	PR after 5 cycles
Max. toxicity	G2 chills/fever
Frequency of symptoms	Acute with each dose
Duration of treatment/ total cycles	8 months/ 7 cycles
Status	Currently on other chemotherapy

The electron microscopy (EM) images from a patient's biopsy of a chest wall mass revealed evidence of viral replication and ghosts/capsids in the patient's tumor . The presence of viral capsids/ghosts is consistent with previous data and reflects the prolonged interval between the first dose of Reolysin and the biopsy (93 days).

TUMOR BIOPSY

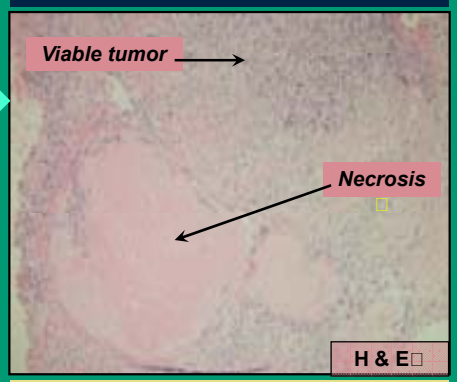
Viable tumor
Necrosis
H & E

Taken 93 days after the 1st dose of Reolysin, (48 hours after the 3rd dose).

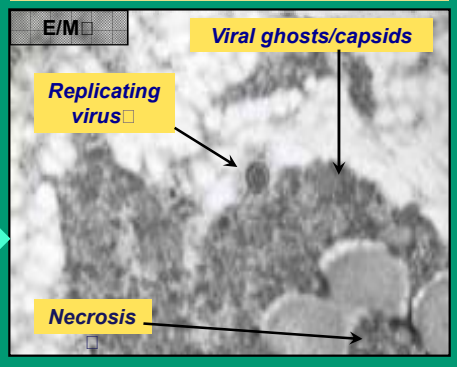
E/M
Viral ghosts/capsids
Replicating virus
Necrosis

COMPUTED TOMOGRAPHY SCAN (CT scan)

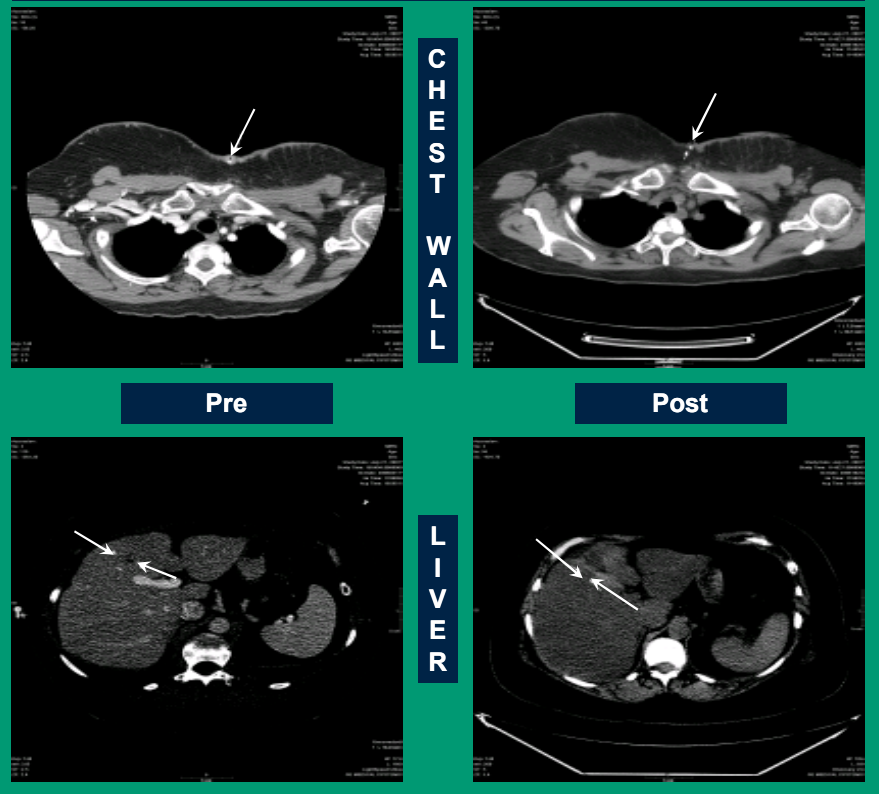
CHEST WALL
Pre
Post
LIVER

RESULTS

MUTATIONAL ANALYSIS

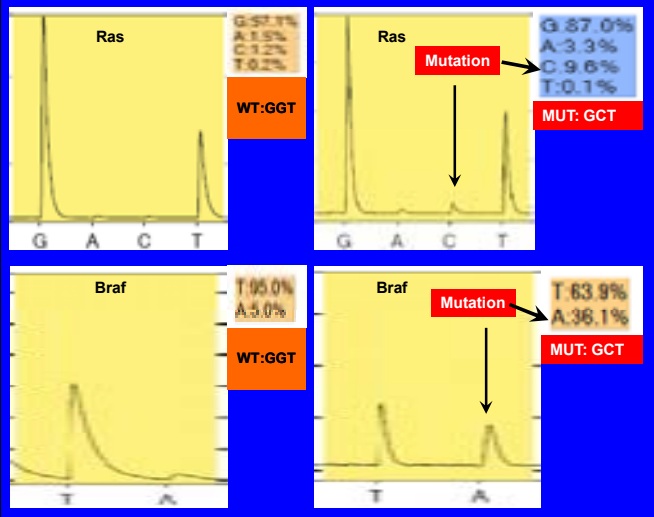
Ras
WT:GGT
Ras
Mutation
G:87.0%
A:3.3%
C:9.6%
T:0.1%
MUT: GCT
Braf
T:95.0%
A:5.0%
WT:GGT
Braf
Mutation
T:63.9%
A:36.1%
MUT: GCT

Seq #	Dose	Primary	#Rx	BR	Mutation status				NA	Viral shedding			
					Ras		Braf			Ser	Sal	Ur	Feces
					12	13	61	600					
101	1	Ovarian	1	SD	WT	WT	WT	WT	D8	N	N	N	N
102	1	Colon	1	PD	WT	WT	WT	WT	D8	N	N	N	N
103	1	Leio*	1	SD	WT	WT	WT	WT	D8	N	N	N	N
201	2	Colon	1	PD	G>V	WT	WT	WT	D8	N	N	N	N
202	2	Ovarian	3	SD	WT	WT	WT	WT	D8	N	N	N	N
203	2	Carcinoid	1	SD	ND	ND	ND	ND	D8	D1	D3	D3	N
301	3	Cervical	1	PD	WT	WT	WT	WT	D5	N	N	N	N
302	3	Ovarian	1	SD	WT	WT	WT	WT	D5	D3	N	D3	N
303	3	Prostate	1	PD	G>A	WT	WT	WT	D28	N	N	N	N
401	4	Ovarian	1	PD	WT	WT	WT	WT	D5	N	N	N	N
402	4	Colon	1	PD	WT	WT	WT	WT	D5	N	N	N	N
403	4	Cervical	1	PD	WT	WT	WT	WT	D8	N	N	N	N
501	5	Breast	7	PR	G>A	WT	WT	WT	D14	N	D149	N	D54
502	5	Breast	1	PD	G>A	WT	WT	V>E	D8	N	N	N	N
503	5	Lung	1	PD	WT	WT	WT	WT	D8	D21	N	D8	N
601	6	Colon	1	PD	WT	WT	WT	WT	D8	N	N	D14	N
602	6	Ovarian	1	SD	WT	WT	WT	WT	D8	N	N	N	N
603	6	Ovarian	2	SD	G>V	WT	WT	WT	D1	D1	D1	D1	D8

VIRAL SHEDDING IN BODILY FLUIDS

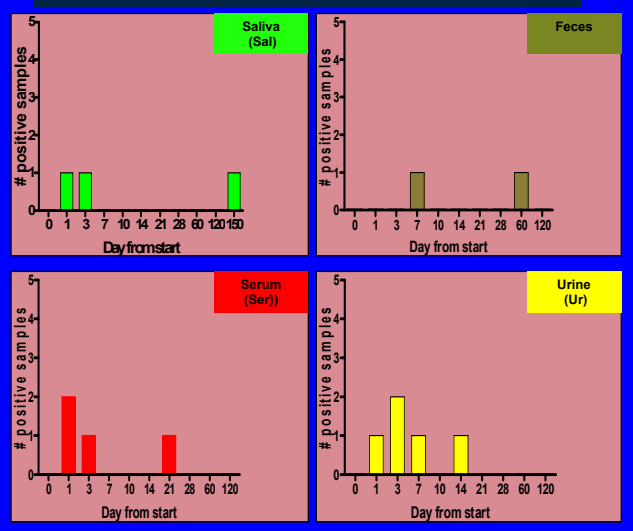
Saliva (Sal)
Feces
Serum (Ser))
Urine (Ur)
positive samples
Day from start
0 1 3 7 10 14 21 28 60 120

NEUTRALIZING ANTIBODY (NA)

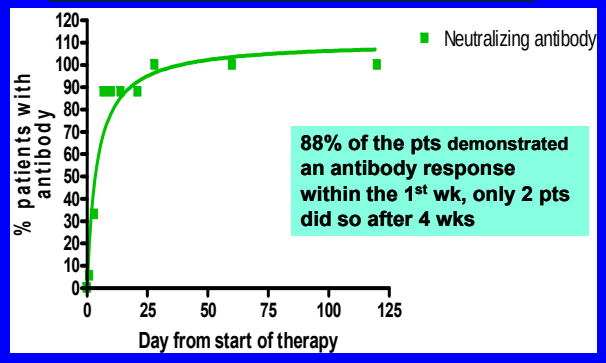
Neutralizing antibody
% patients with antibody
Day from start of therapy
88% of the pts demonstrated an antibody response within the 1st wk, only 2 pts did so after 4 wks

CLINICAL BENEFIT (CB)

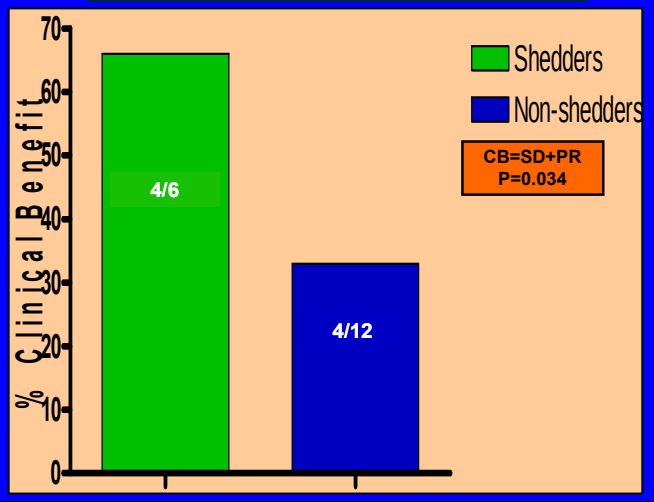
Shedders
Non-shedders
CB=SD+PR
P=0.034
4/6
4/12
% Clinical Benefit

TOXICITY & RESPONSE

Toxicity	GRADE I	GRADE II
Fever	5	1
Chills	-	-
Fatigue	4	1
Diarrhea	4	-
Nausea	5	-
Vomiting	-	-
Headache	5	-

Response & DLT	PR	SD	DLT
	1 34% by RECIST	7	None

CONCLUSIONS

- Reolysin at 3X10^{10} TCID$_{50}$ q 4 wk IV is safe and well tolerated
- Grade 2 toxicity is manageable
- Clinical activity in refractory patients
- Warrants phase II testing (single agent sarcoma trial initiated)
- Viral shedding possibly portends better response
- Role of Ras mutational analysis?